Dual targeting of ALDH1 and ALDH3:
A promising therapeutic approach in acute myeloid leukemia

Julien Colle[1], Muhube Yazir[2], Céline Baier[2], Yasmine Labiad[2], Isabelle Arnoux[3], Geoffroy Venton[1], Guillaume Martin[2], Ismail Ceylan[2], Régis Costello[1], Mileidys Perez-Alea[2]

1- CHU de la Conception, Hématologie et Thérapie Cellulaire, 147 Boulevard Baille, 13005 Marseille *2- Advanced BioDesign ABD, Parc Technologique de Lyon, 655 Allée des Parcs, 69800 Saint-Priest*

Background

The aldehyde dehydrogenases family *(ALDHs)* play a major cytoprotective role in cells via detoxification of a wide range of aldehydes. Their functional activity also extends to modulation of cell proliferation, differentiation, drug resistance and survival of cancer cells. Despite evidences suggesting that ALDHs could be a promising target for cancer treatment, the identity of the specific isoforms contributing to this in acute myeloid leukemia (AML), the underlying mechanisms and the therapeutic value of ALDH inhibitors remain elusive.

Methods

We conducted a meta-analysis to investigate the impact of ALDHs on clinicopathological parameters and survival in AML patients. In an independent cohort (N=110), the expression of ALDHs and their corresponding enzymatic activities were monitored in AML patient-derived cells and pharmacologically inhibited with DIMATE, an irreversible suicide inhibitor of ALDH1 and ALDH3. Targeting of ALDH was evaluated both, *in vitro* and in orthotopic xenograft tumor models and the molecular mechanisms associated were studied in loss-of-function experiments, luciferase reporter assays and combination-index for DIMATE-drug interactions.

Results

Genes coding for ALDHs class 1&3 are up-regulated in about 45% of AML patients collected in the TCGA database (N = 200). These genes present a mutually exclusive pattern, suggesting that they may have similar functions. The high expression of these isoenzymes revealed shorter survival times in patients and strong impact on chemotherapy resistance.

The Cancer Genome Atlas (TCGA)

Genes altered in 45% of patients (N=200)

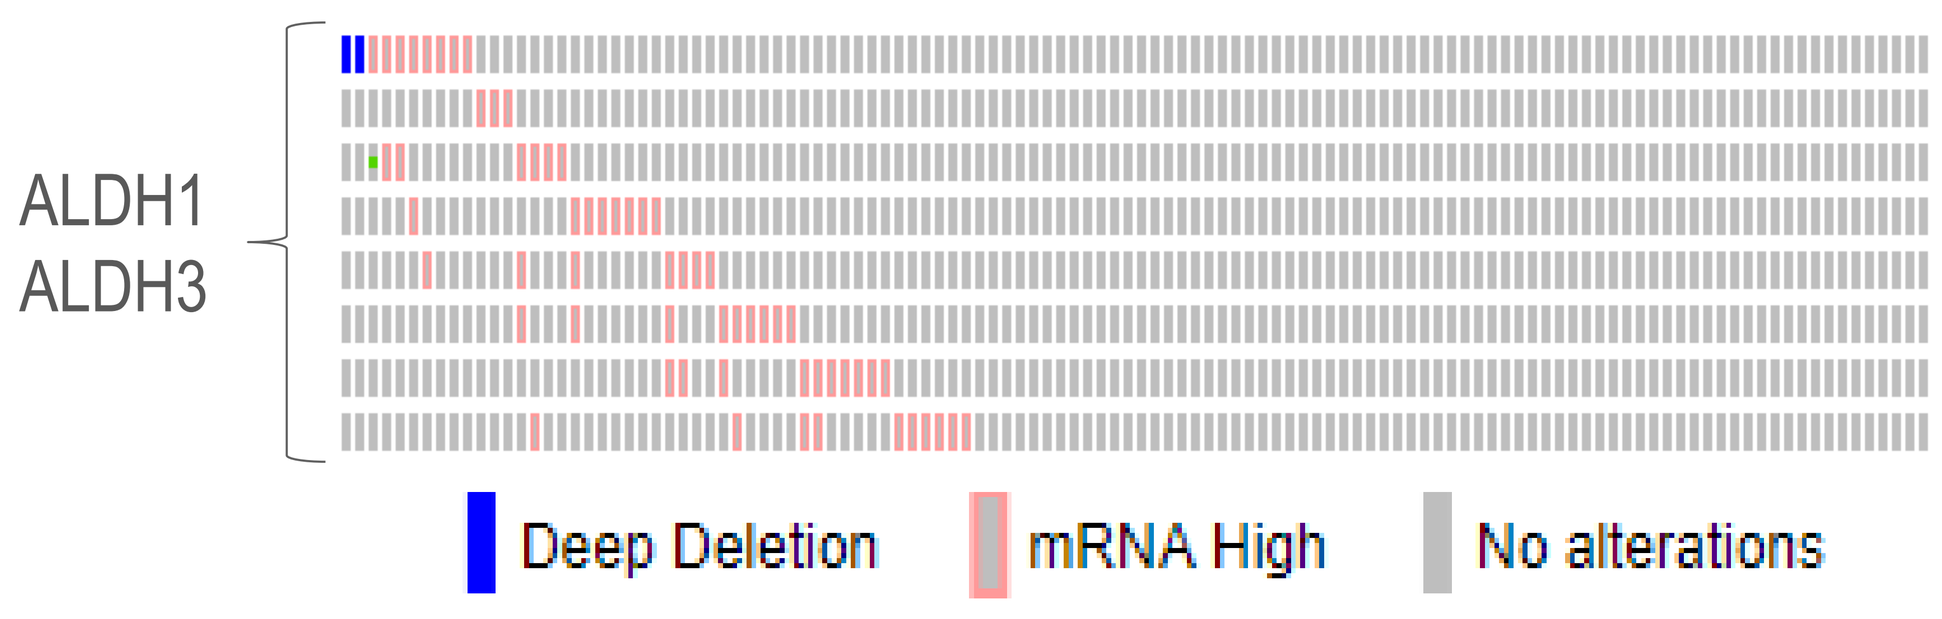
ALDH1
ALDH3
Deep Deletion
mRNA High
No alterations

Worse overall survival in patients with high ALDH activity

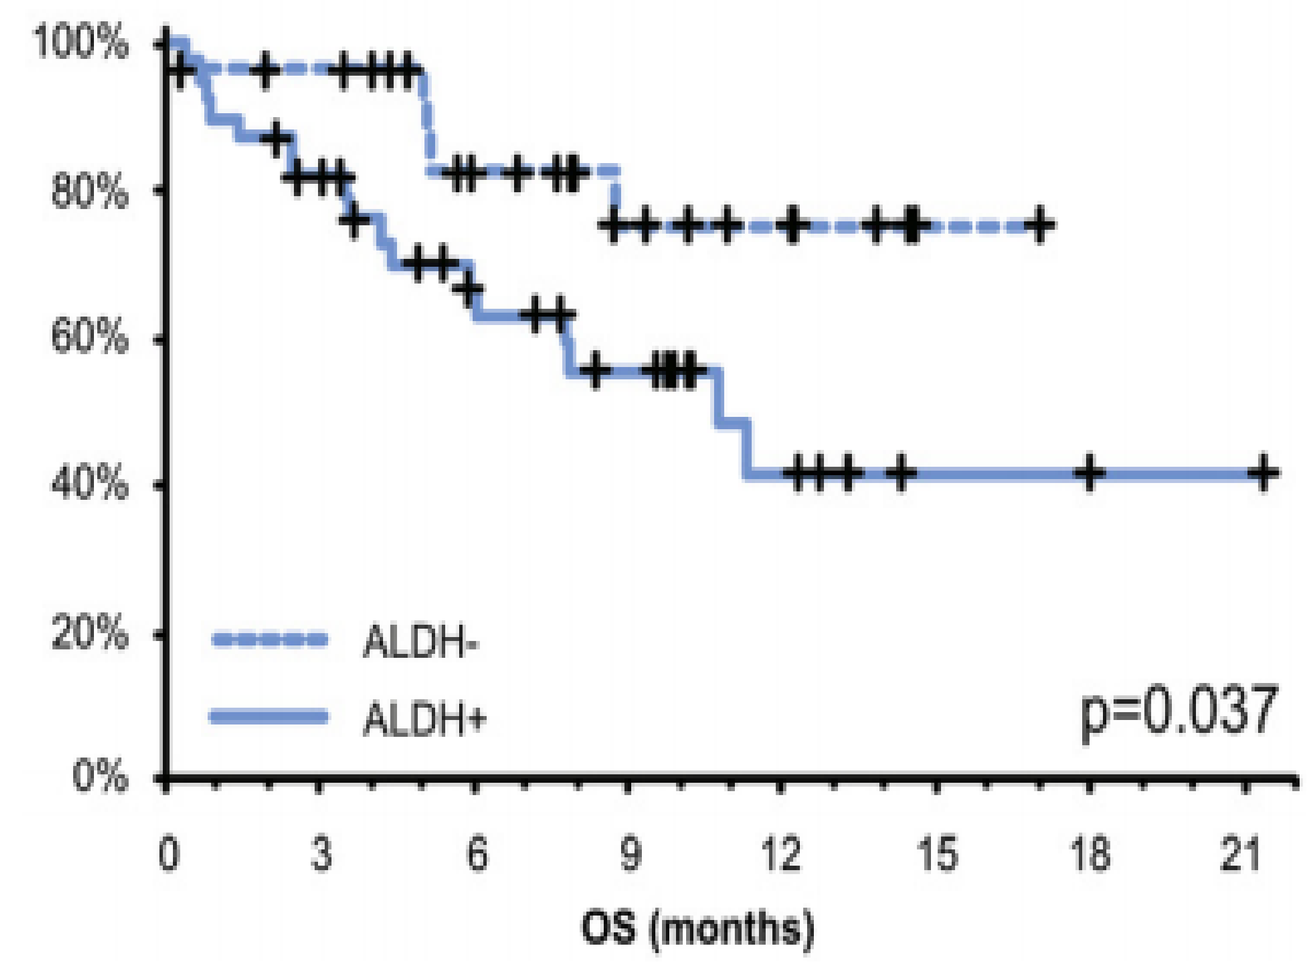
100%
80%
60%
40%
20%
0%
ALDH-
ALDH+
p=0.037
0 3 6 9 12 15 18 21
OS (months)

The subgroup of patient with the highest levels of ALDH1 have the worse prognosis. In addition, ALDH1 activity was significantly higher in refractory patients and in the subgroup of patients with adverse prognosis, according to the ELN molecular/ cytogenetic classification of 2017 (p<0.005)

ALDH activity is higher in refractory vs non-refractory AML (N=116)

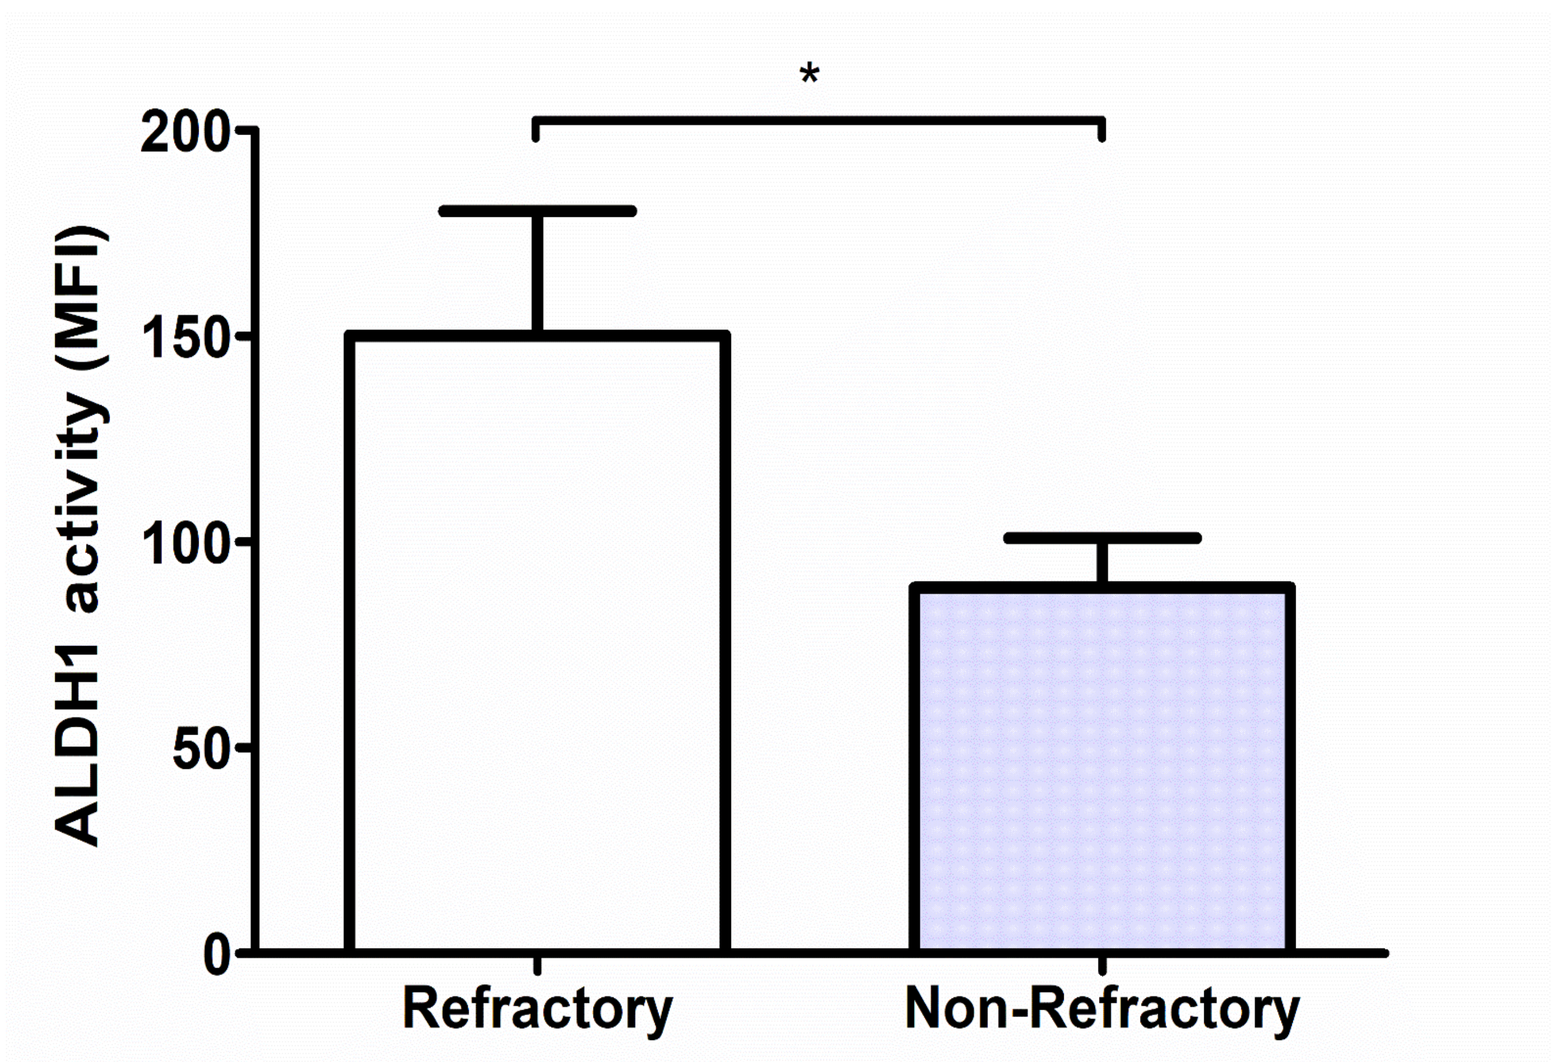
ALDH1 activity (MFI)
Refractory
Non-Refractory

Patients with adverse prognosis present higher ALDH activity than subgroups with intermediate and favourable risk factors (N= 116; p<0.005)

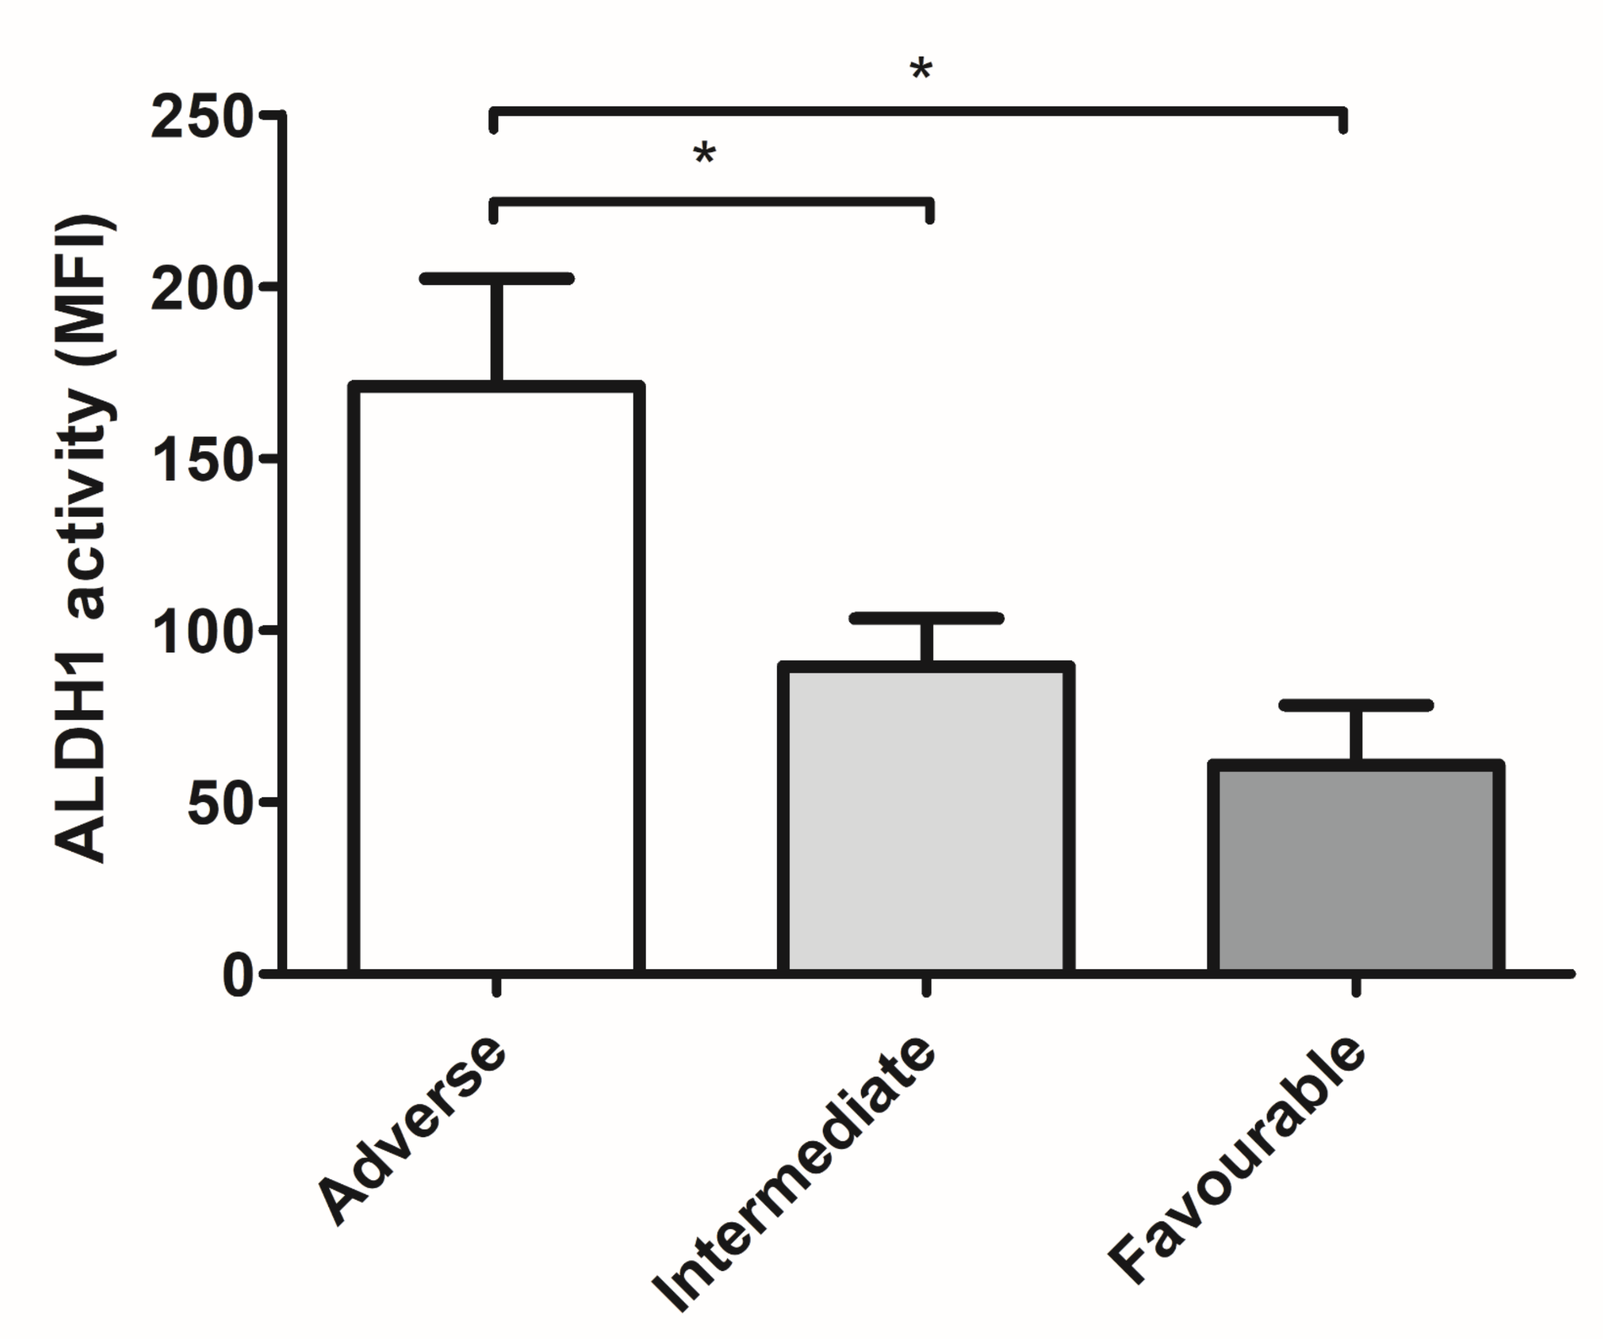
ALDH1 activity (MFI)
Adverse
Intermediate
Favourable

ALDH 1 activity in blast cells (N= 65)

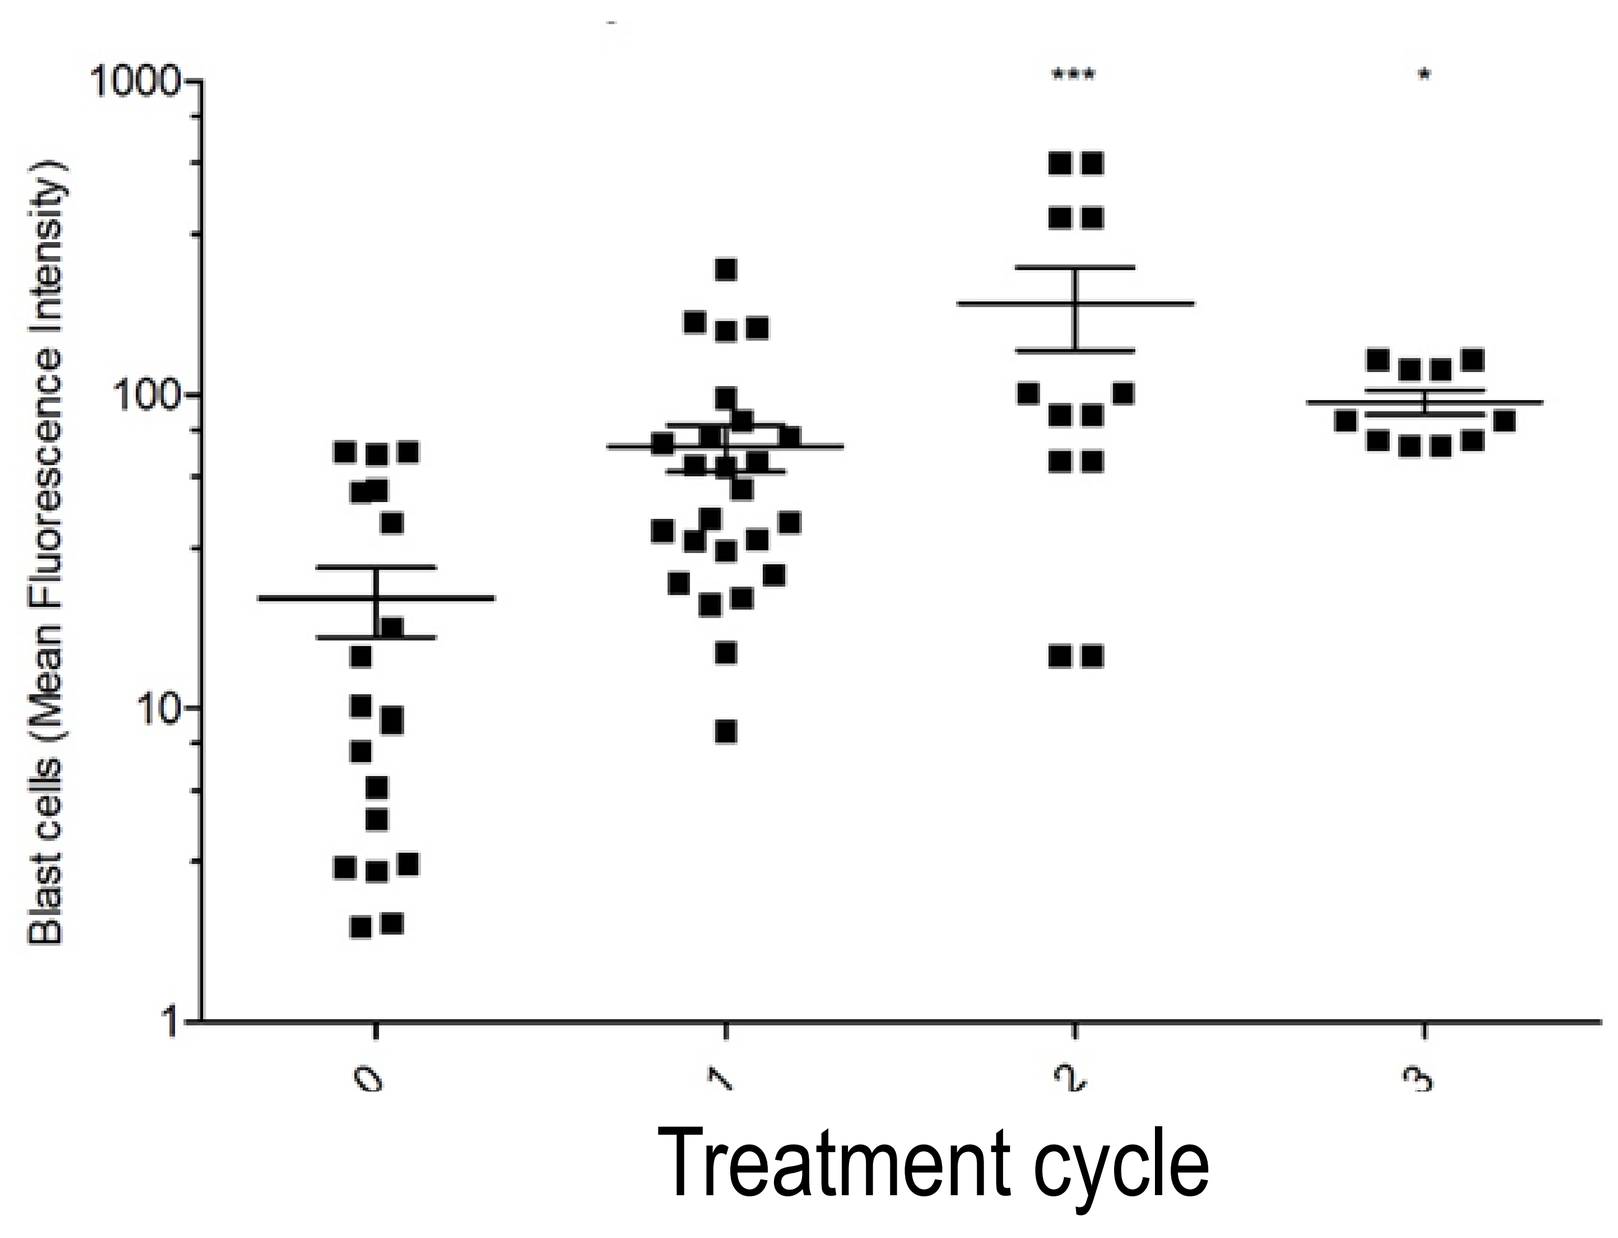
Blast cells (Mean Fluorescence Intensity)
Treatment cycle

In a prospective cohort (N=110), as patients underwent different cycles of classical chemotherapeutic regimen with daunorubicine (DNR) and/or Cytarabine (AraC), the remaining blasts were increasingly enriched for ALDH1 activity.

In vitro and *in vivo* studies

Whether the increasing enrichment of ALDH-high blasts in treated patients is due to the initial existence of drug-resistant, ALDH-high clones or the adaptative response to the drug-selective pressure remains to be clarify

Using commercially available AML cells lines we found that three specific isoforms of the ALDH class 1& 3 subfamilies are upregulated following *in vitro* treatment with cytarabine (Ara-C) or daunorubicin (DNR).

ALDH 1 &3 up-regulation in response to AraC and Dauno

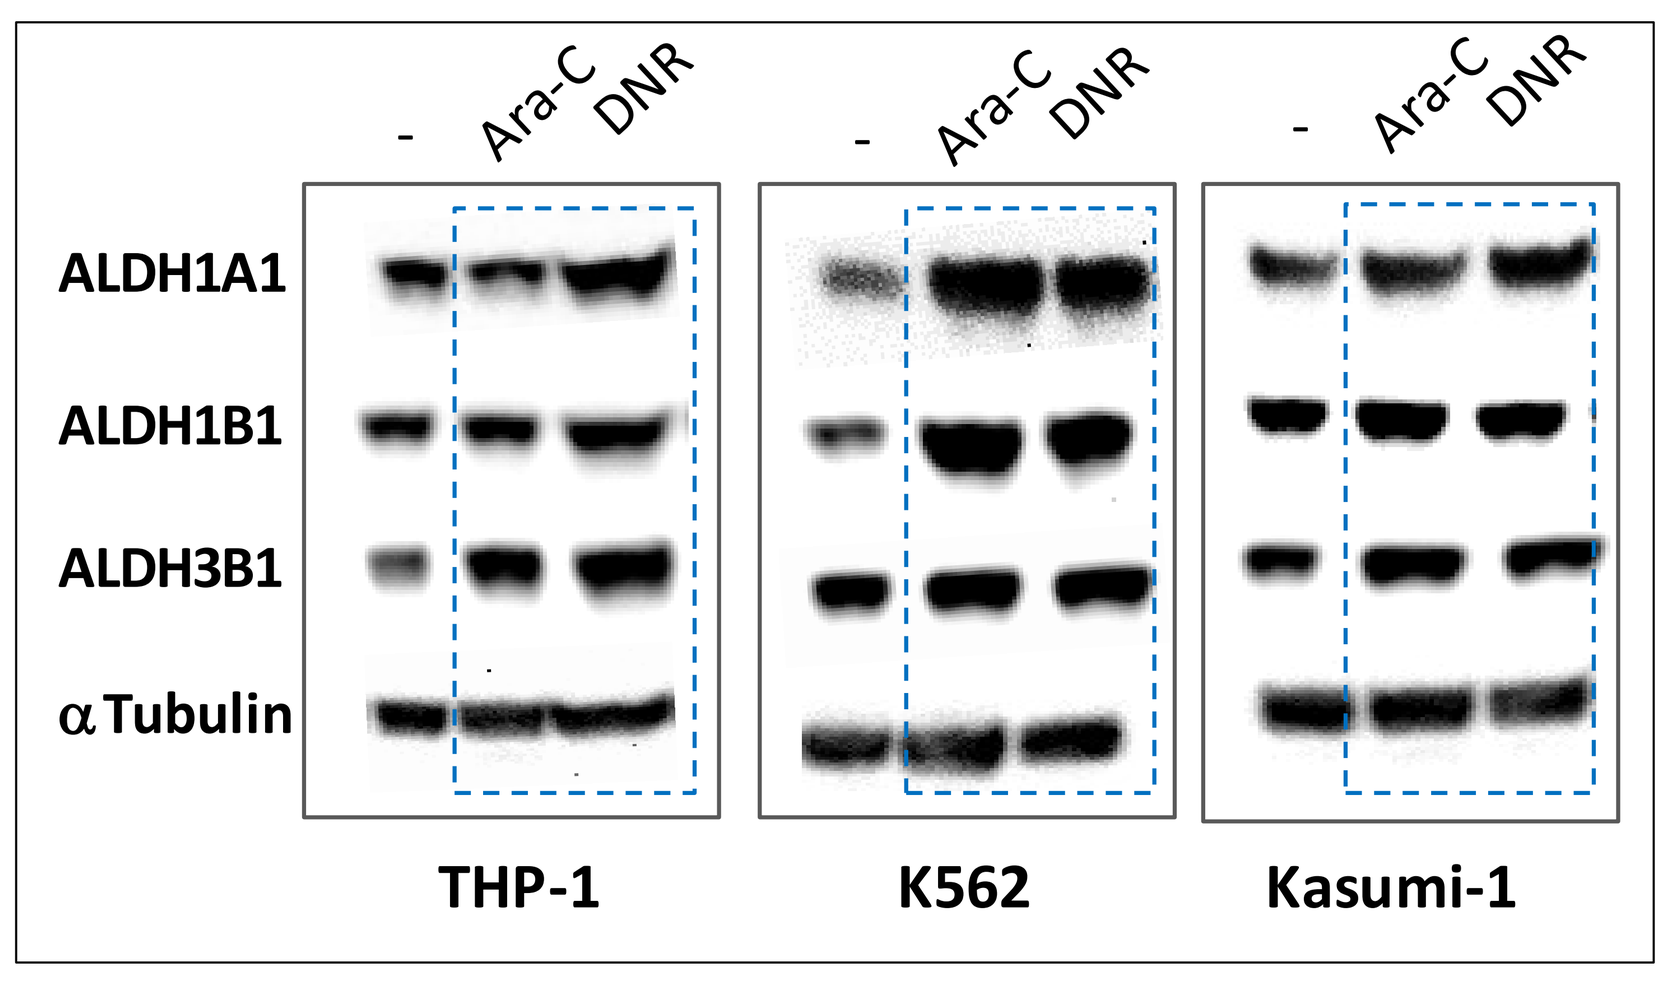
- Ara-C DNR
ALDH1A1
ALDH1B1
ALDH3B1
αTubulin
THP-1
K562
Kasumi-1

Higher cytotoxic effect of the ALDH1&3 inhibitor DIMATE in AML blast vs normal haematopoietic CD34+ cells

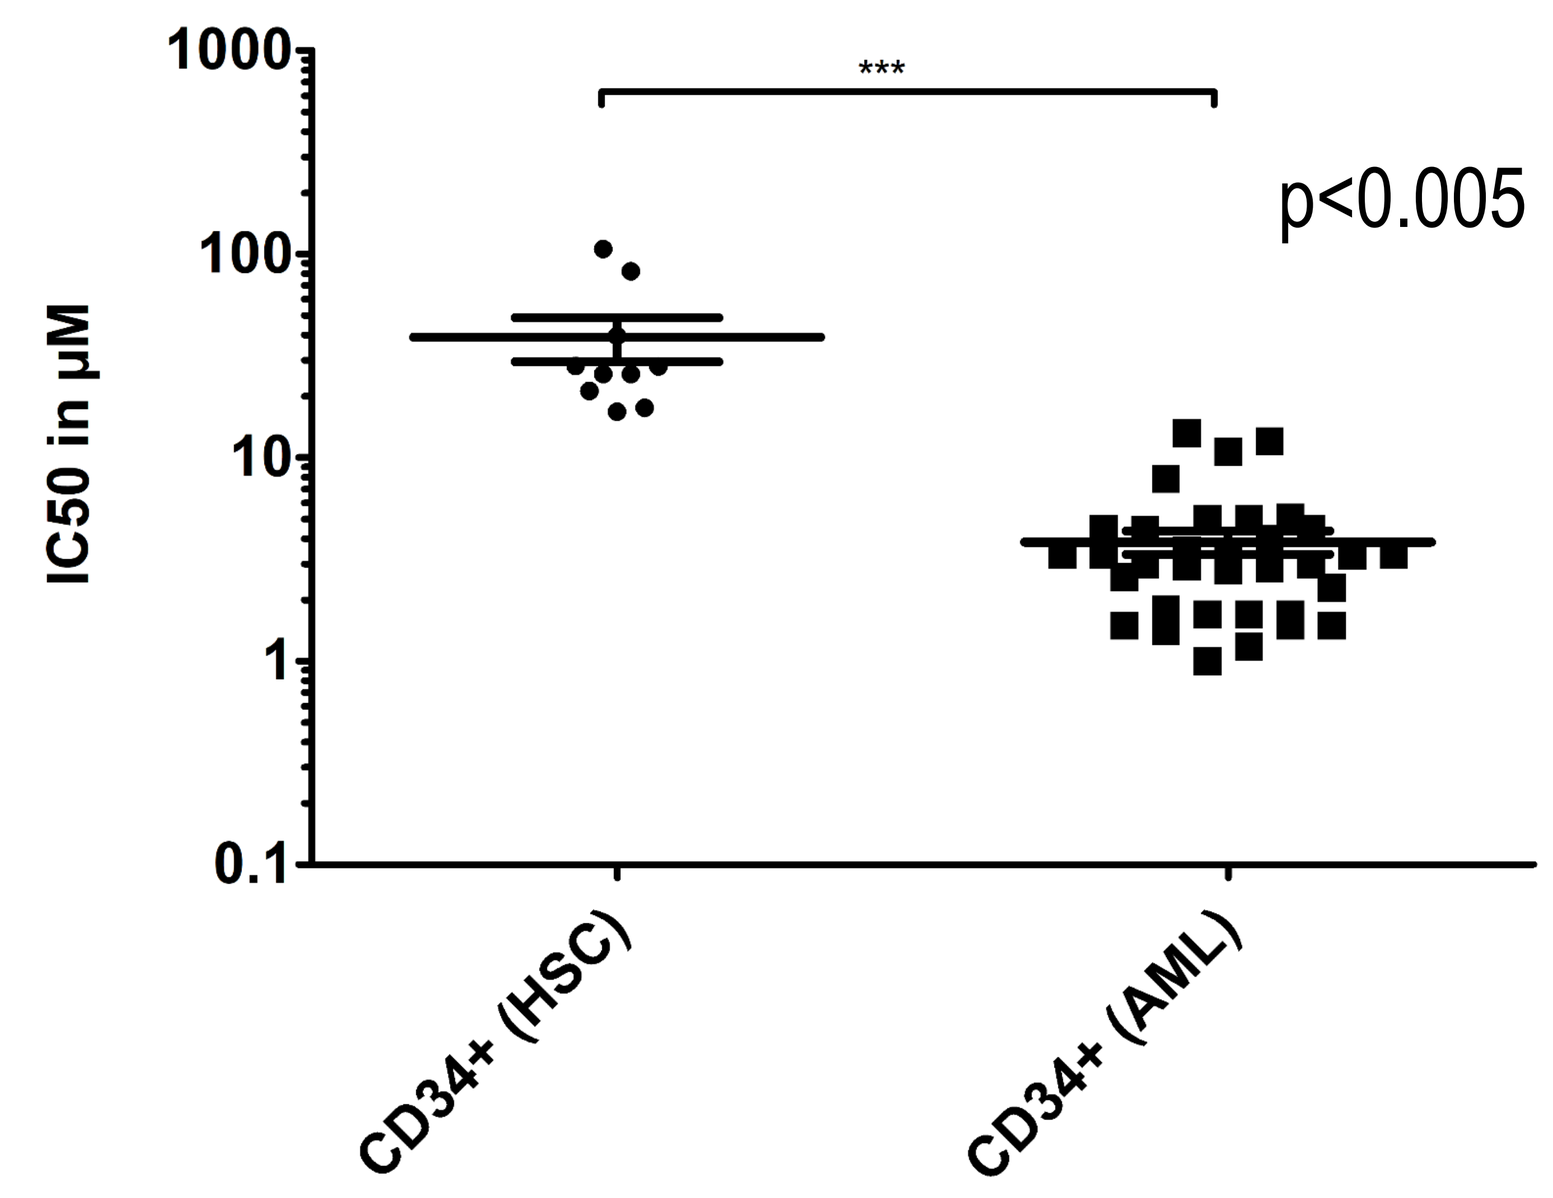
IC50 in μM
p<0.005
CD34+ (HSC)
CD34+ (AML)

Representation of genes that are modified in response to DIMATE in AML cell lines, including *CHAC1 & DDIT3* ferroptosis-associated genes

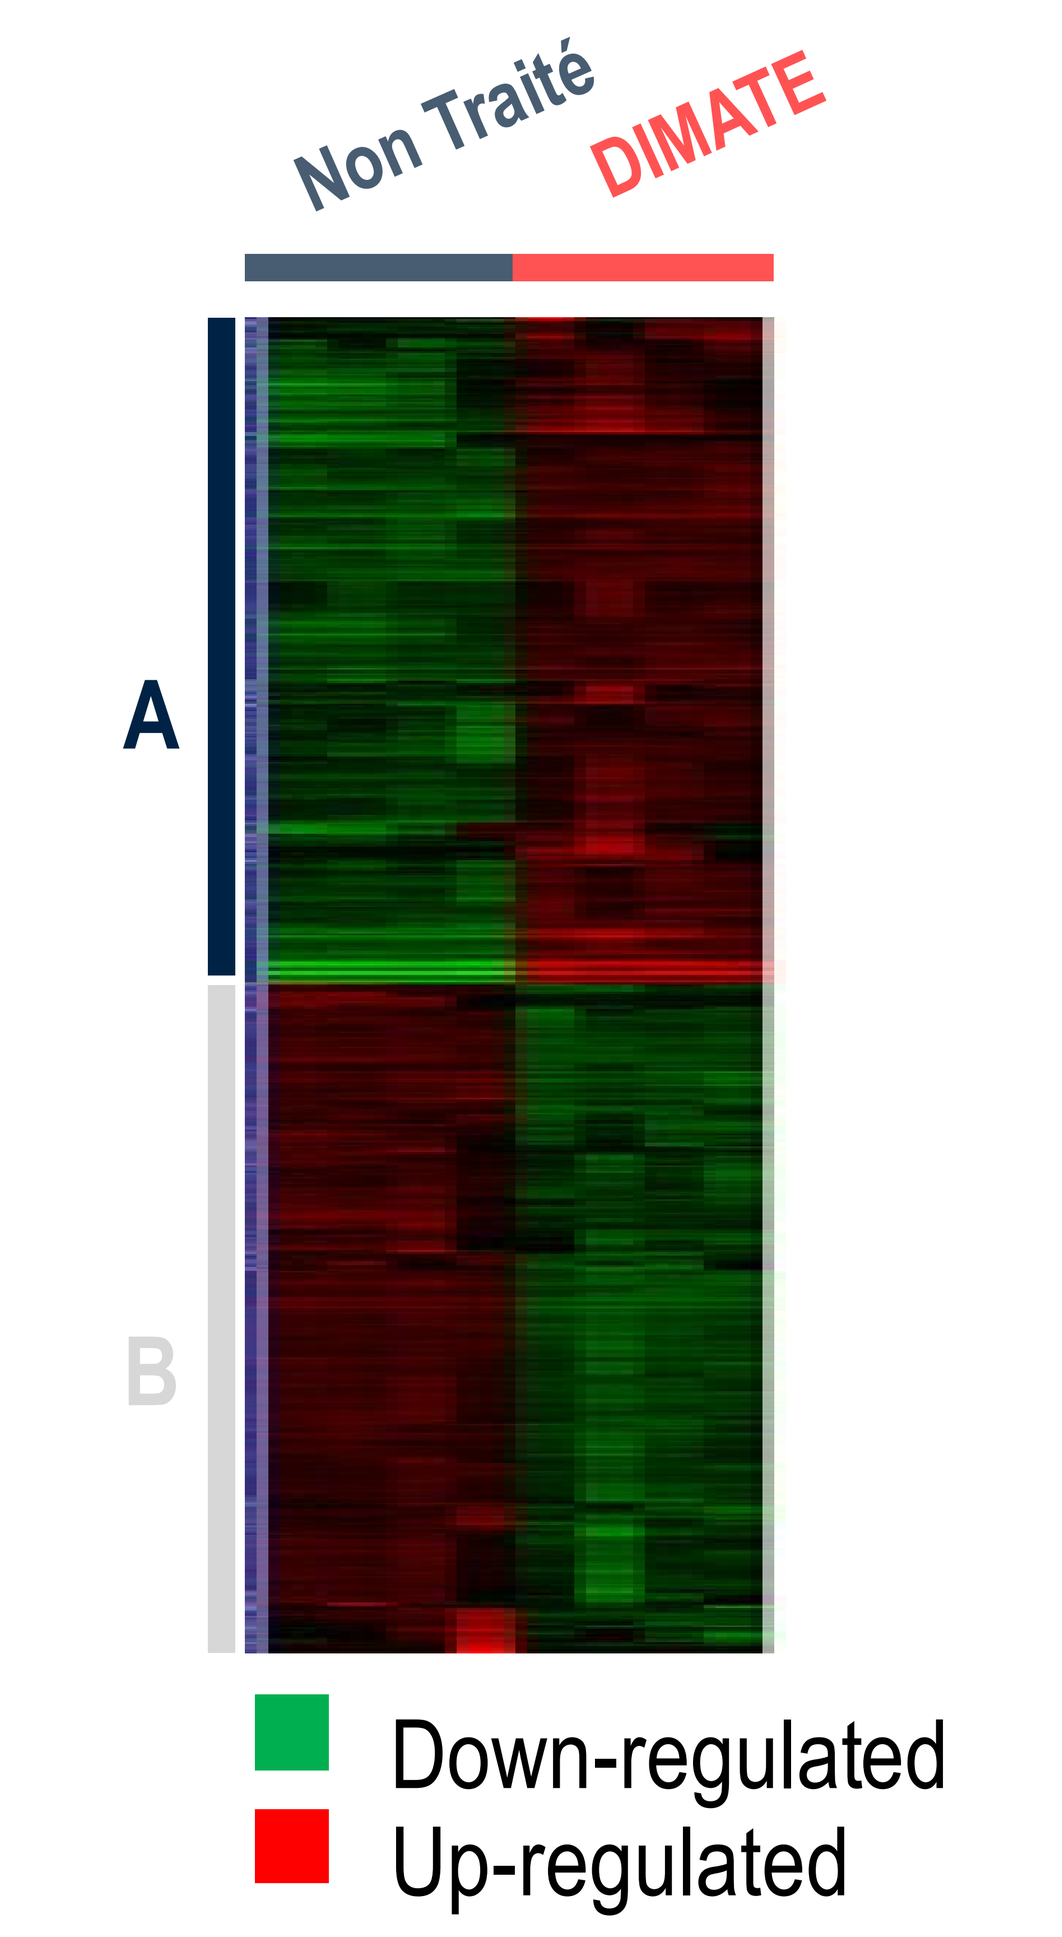
Non Traité
DIMATE
A
B
Down-regulated
Up-regulated

Activation of JNK stress pathway in response to DIMATE

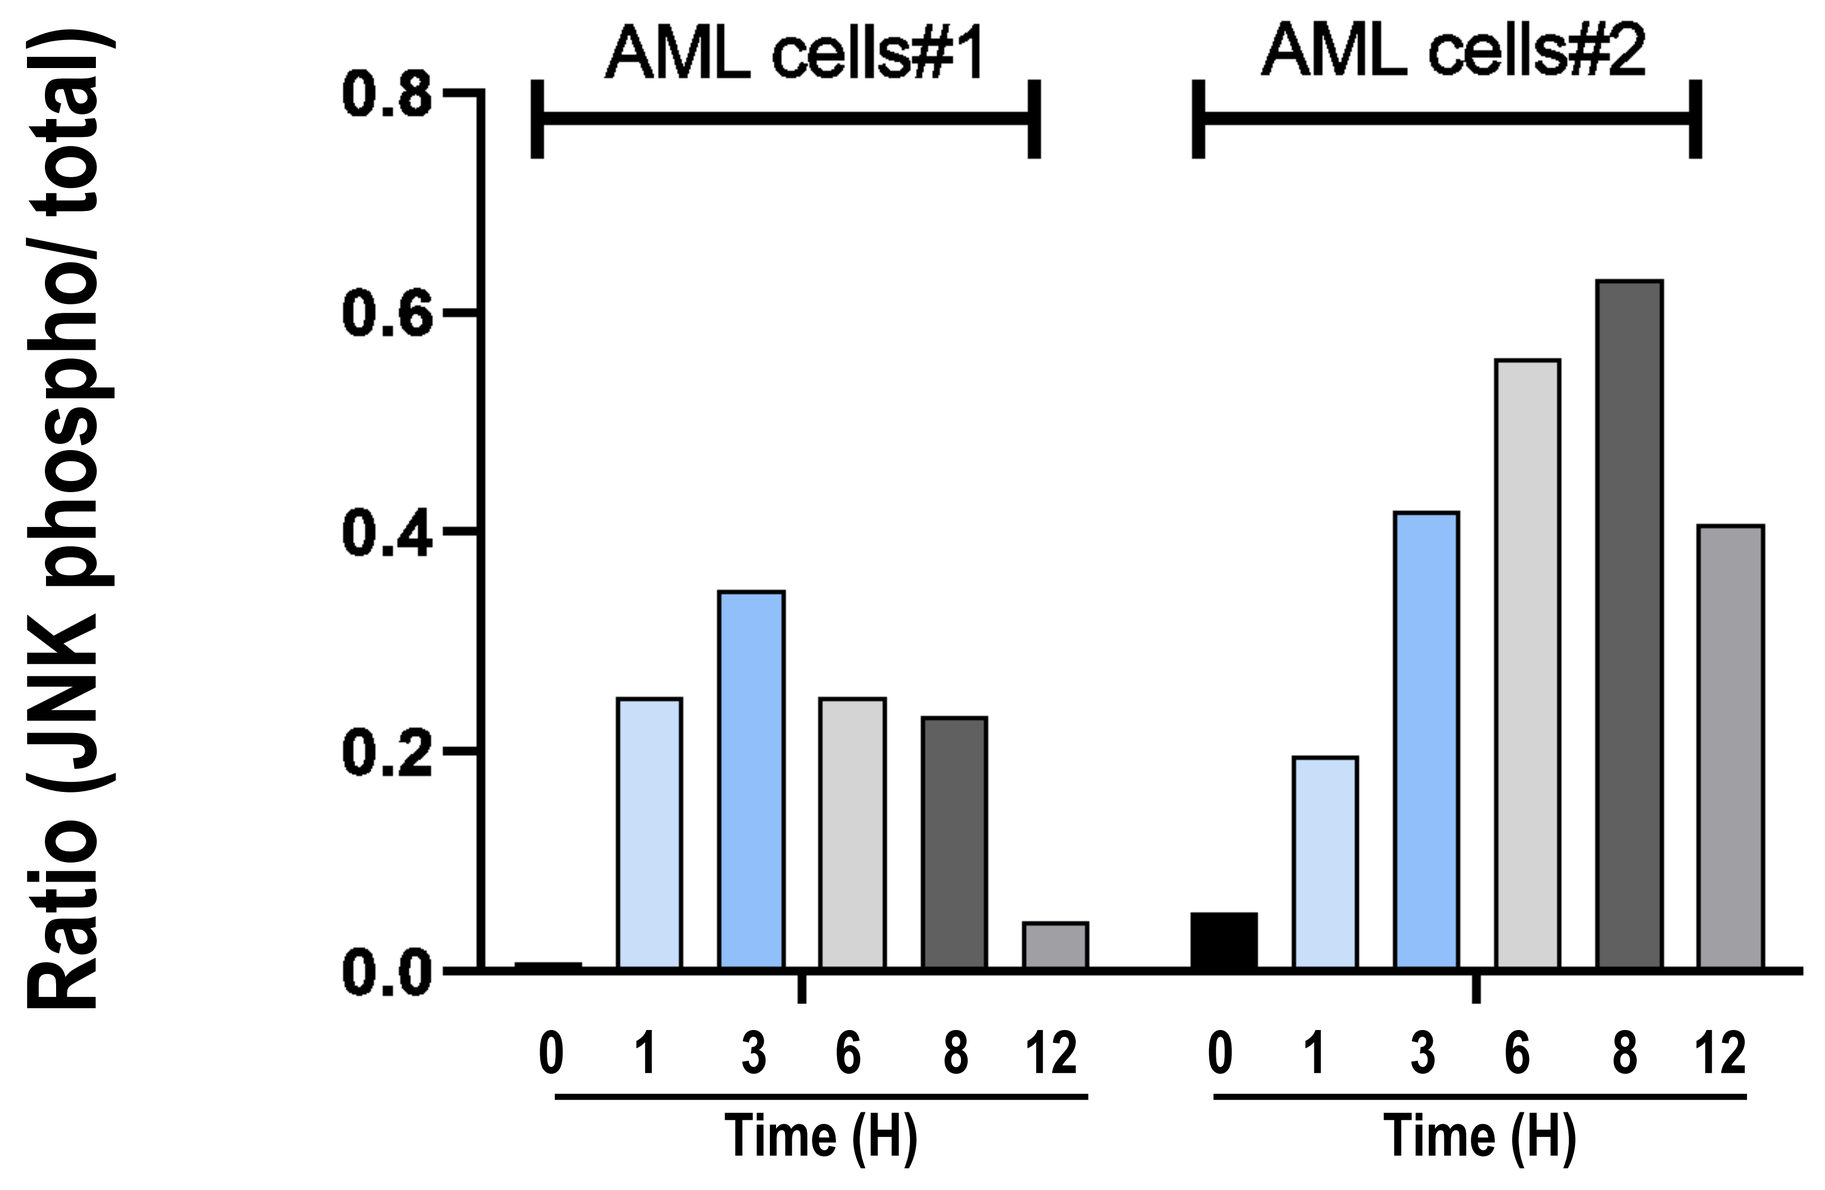
Ratio (JNK phospho/ total)
AML cells#1
AML cells#2
0 1 3 6 8 12
Time (H)

Antitumor effect of DIMATE in xenograft mice, generated from CD34+ leukemic cells*

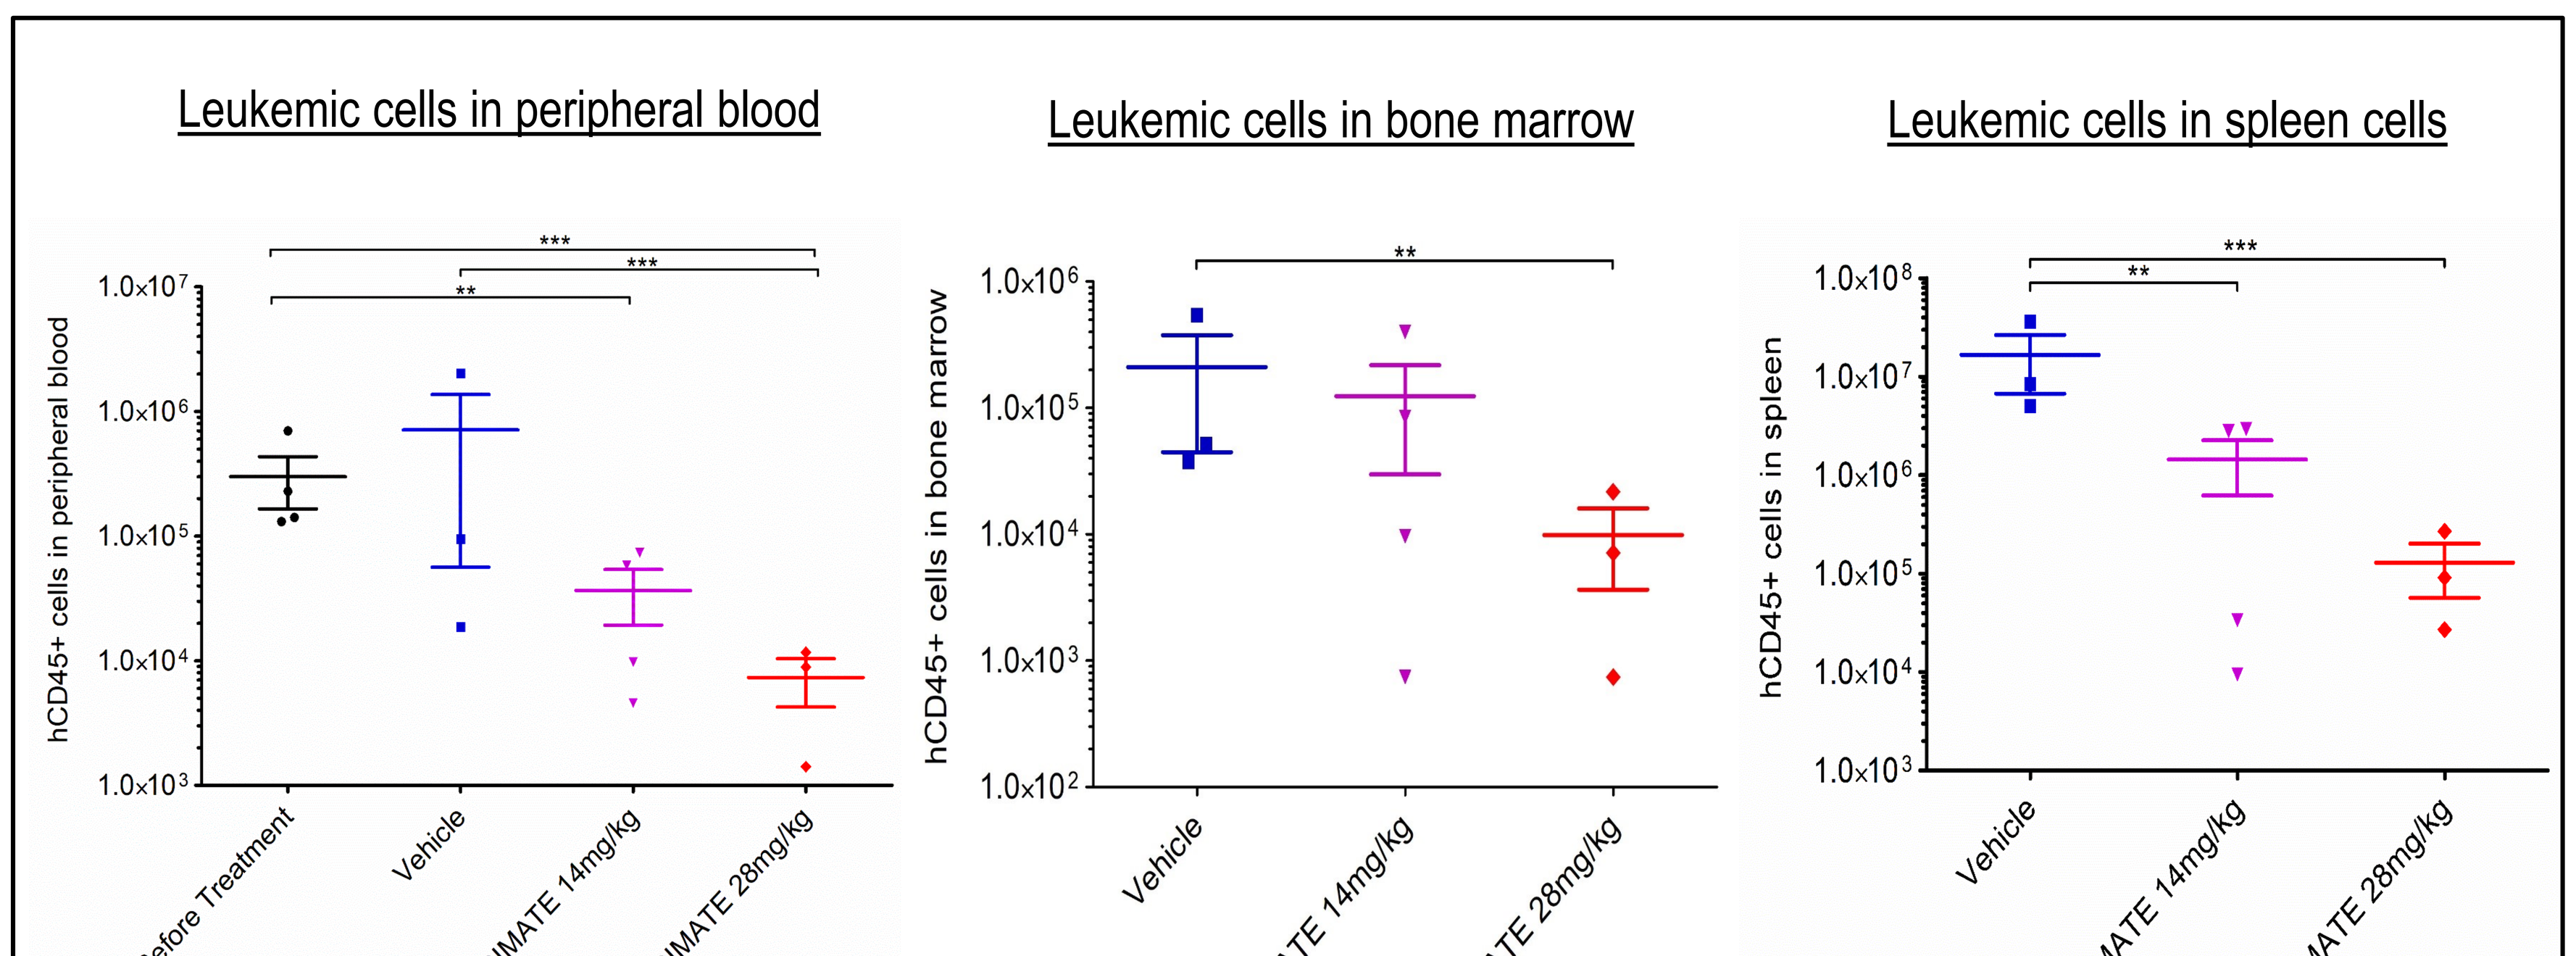
Leukemic cells in peripheral blood
Leukemic cells in bone marrow
Leukemic cells in spleen cells
hCD45+ cells in peripheral blood
hCD45+ cells in bone marrow
hCD45+ cells in spleen
Before Treatment
Vehicle
DIMATE 14mg/kg
DIMATE 28mg/kg

** CD34+ leukemic PBMC were from a patient with refractory AML (complex karyotype+del(5q)), poor prognosis, after induction therapy*

Intraperitoneal administration of DIMATE (3twk) induced a significant depletion of human leukemic cells in peripheral blood, as well as in bone marrow and spleen organs in all the animals receiving the doses of 28mg/kg of the drug.

Conclusion

Our data provide a proof of concept that AML with increased expression of ALDH1 and 3 may benefit from strategies including inhibitors of these isoenzymes, as single agents or in combination with chemotherapy to overcome patient-specific drug resistance. For these patients, who often have poor responses and no alternative therapeutic plan, DIMATE could be a promising option.

Bibliography

G Venton, Blood Cancer J. 2016; M Perez-Alea, Oncogene. 2017; R. Rebollidos Oncogene. 2019